Multiple Modes of Action of the CD38 x ICAM-1 Bispecific Antibody

Xiaocheng Chen, Oi Kwan Wong and Leonard Post

Virtuoso Therapeutics, San Mateo, California, USA

AACR #5385

Background

Targeting cluster of differentiation 38 (CD38) with monoclonal antibodies has resulted in outstanding responses in patients with multiple myeloma (MM). However, a significant portion of patients failed to respond and nearly all patients eventually relapsed. One of the mechanisms of resistance has been partially attributed to lower CD38 expression. Furthermore, daratumumab, an anti-CD38 antibody, only showed limited monotherapy activity in relapsed/refractory Non-Hodgkin lymphoma (NHL) patients in a phase 2 study. Therefore, there are opportunities for second generation CD38 based therapies. We hypothesize that a bispecific antibody with one arm targeting a tumor associated guide antigen (TAG) and the other arm targeting CD38 could achieve tumor specificity and improve potency[1].

Intercellular Adhesion Molecule 1 (ICAM-1), an Immunoglobulin (Ig)-like Cell Adhesion Molecule, is Highly Expressed in Multiple Myeloma and Lymphoma

The intercellular adhesion molecule-1 (ICAM-1, CD54) is a type I transmembrane glycoprotein and a ligand for leukocyte integrins a_Lb_2 (LFA-1, CD11a/CD18) and a_Mb_2 (Mac-1, CD11b/CD18). ICAM-1 is constitutively present at low levels on endothelial cells, but its overexpression has been observed on various types of tumors and the associated stroma[2]. ICAM-1 is highly expressed on cell surface of primary myeloma cells[3]. Antibody against ICAM-1, Bersanlimab (BI505, BioInvent), was well-tolerated, but only showed limited clinical efficacy in MM patients[4].

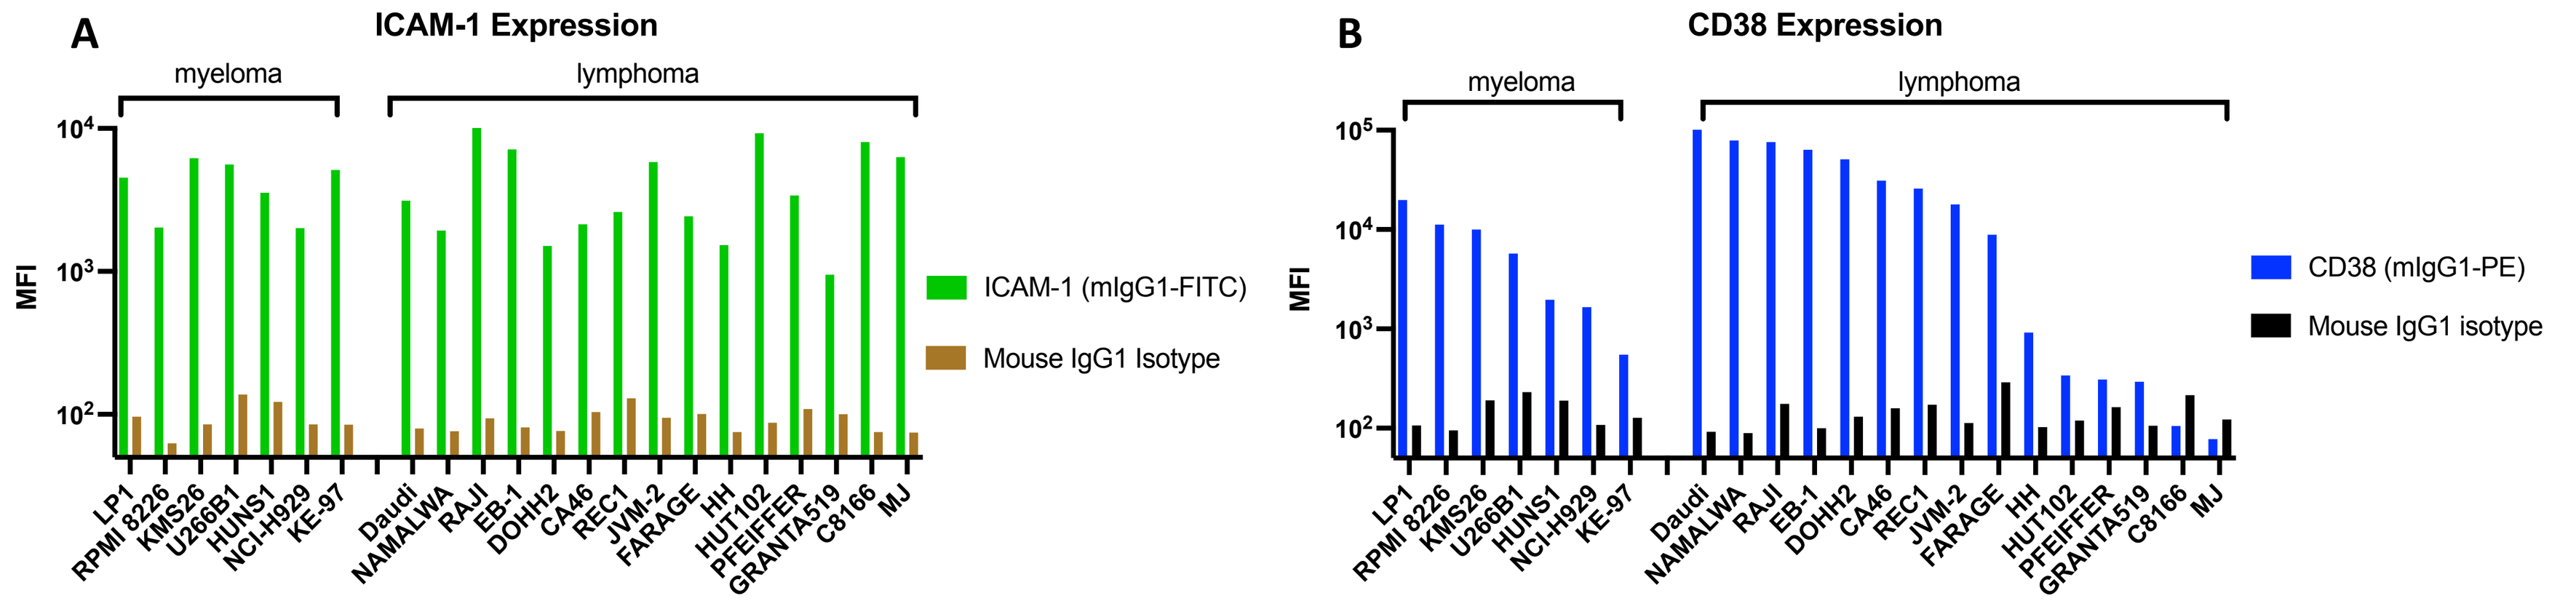
A
ICAM-1 Expression
myeloma
lymphoma
MFI
ICAM-1 (mIgG1-FITC)
Mouse IgG1 Isotype
B
CD38 Expression
CD38 (mIgG1-PE)
Mouse IgG1 isotype
LP1
RPMI 8226
KMS26
U266B1
HUNS1
NCI-H929
KE-97
Daudi
NAMALWA
RAJI
EB-1
DOHH2
CA46
REC1
JVM-2
FARAGE
HH
HUT102
PFEIFFER
GRANTA519
C8166
MJ

Fig 1. ICAM-1 and CD38 expression levels were evaluated using flow cytometry. ICAM-1 is highly expressed in a selected set of myeloma and lymphoma cell lines **(A)**, whereas these cell lines have a range of different CD38 expression **(B)**. Detection antibodies: Biolegend anti-human CD38 (Cat #303506) and anti-human ICAM-1 (Cat #322714).

VP301, a Novel ADCC Enhanced CD38 x ICAM-1 Bispecific Antibody

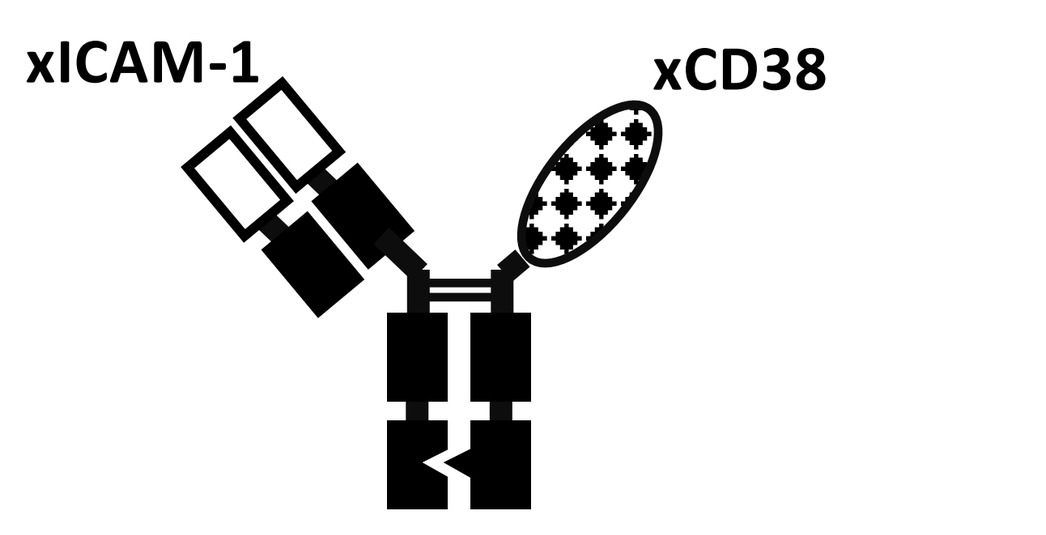
xICAM-1
xCD38

Fig 2. Schematic structure of VP301

Anti-CD38 arm:
❖Proprietary antibody
❖Cross binding to human and cyno CD38 with double-digit nM affinity

Anti-ICAM-1 arm:
❖Proprietary antibody
❖Cross binding to human and cyno ICAM-1 with sub nM affinity

Fc:
❖ Afucosylated human IgG1 antibody with knob-into-hole technology

VP301 Binds to ICAM1 D4D5 Domain, Away from LFA1 Binding Region

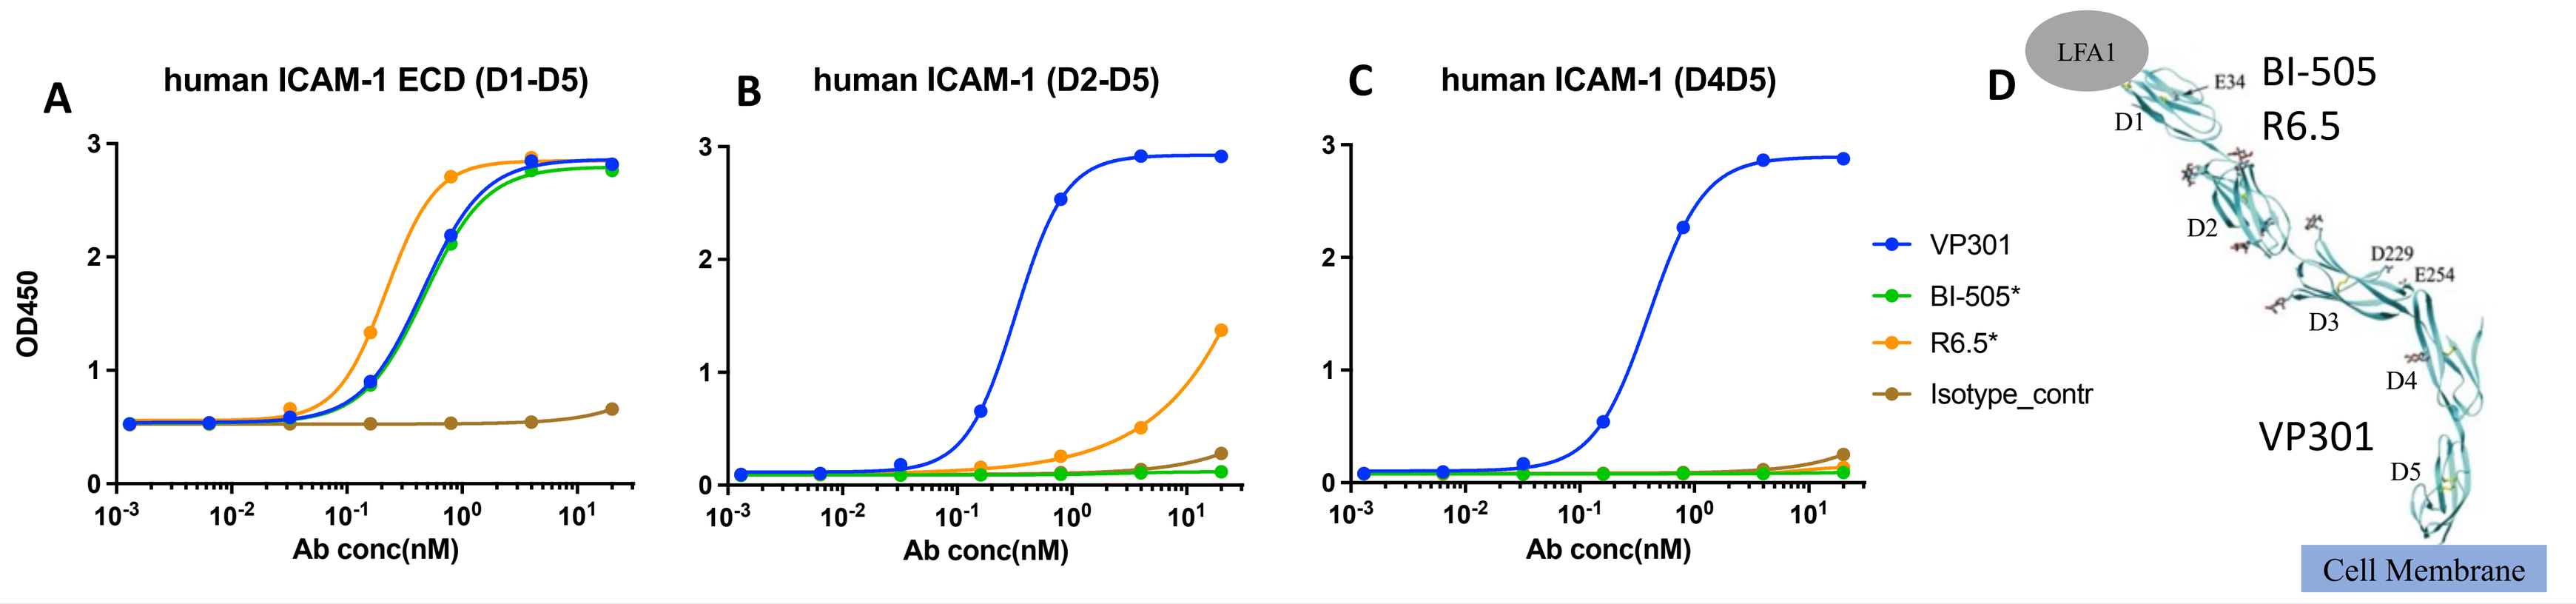
A
human ICAM-1 ECD (D1-D5)
B
human ICAM-1 (D2-D5)
C
human ICAM-1 (D4D5)
OD450
Ab conc(nM)
VP301
BI-505*
R6.5*
Isotype_contr
D
LFA1
BI-505
R6.5
VP301
Cell Membrane

Fig 3. ELISA binding of VP301 to human ICAM-1 ECD domains in comparison to the benchmark anti-ICAM-1 antibodies BI-505 and R6.5. **(A)** All three antibodies showed potent binding to the full length ECD domain. **(B)** VP301 retained good binding to the D1 truncated ICAM-1 regions, whereas R6.5 showed much reduced binding and BI-505 completely abolished the binding. **(C)** VP301 retained good binding to ICAM1 D4D5 domain, whereas neither R6.5 nor BI-505 showed any detectable binding. **(D)** The modeled ICAM-1 ECD structure with antibody binding regions illustrated. *The benchmark anti-ICAM-1 antibodies BI-505 and R6.5 (Enlimomab) were produced by transient expression in CHO cells based on the published sequence information.

VP301 Binds to a Unique Epitope on CD38, and Showed Minimal Binding to Human Red Blood Cells (RBCs)

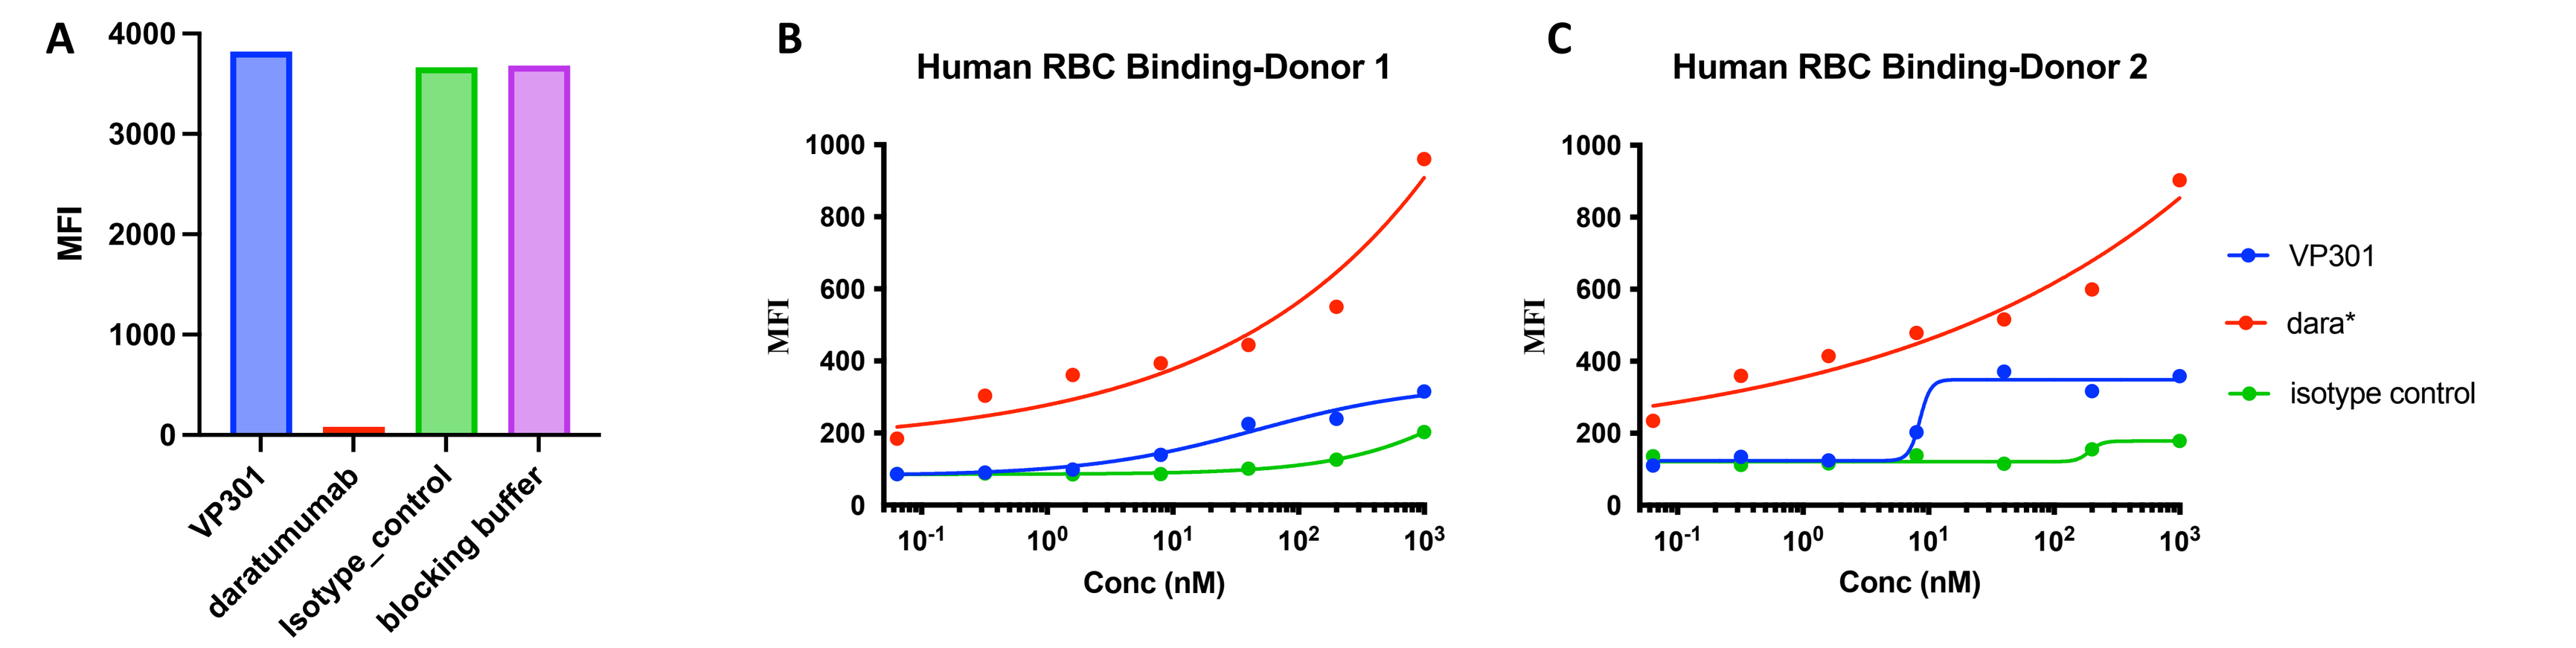
A
MFI
VP301
daratumumab
Isotype_control
blocking buffer
B
Human RBC Binding-Donor 1
C
Human RBC Binding-Donor 2
Conc (nM)
VP301
dara*
isotype control

Fig 4. (A) FACS based epitope binning showed that VP301 does not interfere with dara binding to Raji cells. The biotin labelled dara were incubated with Raji cells in the absence or presence of 100nM of VP301 or control antibodies. As expected, daratumumab (purchased from J&J) strongly blocked dara binding. **(B and C)** VP301 showed much weaker RBC binding compared to dara. Human RBCs from two different donors were incubated with serial dilutions antibodies (from 1uM) at 4℃ for 1 h. The binding was evaluated using flow cytometry. *The anti-CD38 benchmark antibody dara was produced by transient expression in CHO cells based on the published sequence information for the commercially marketed anti-CD38 antibody, daratumumab.

VP301 Showed Potent Binding to Human Fc-gamma Receptor IIIa (FcγRIIIa;CD16a)

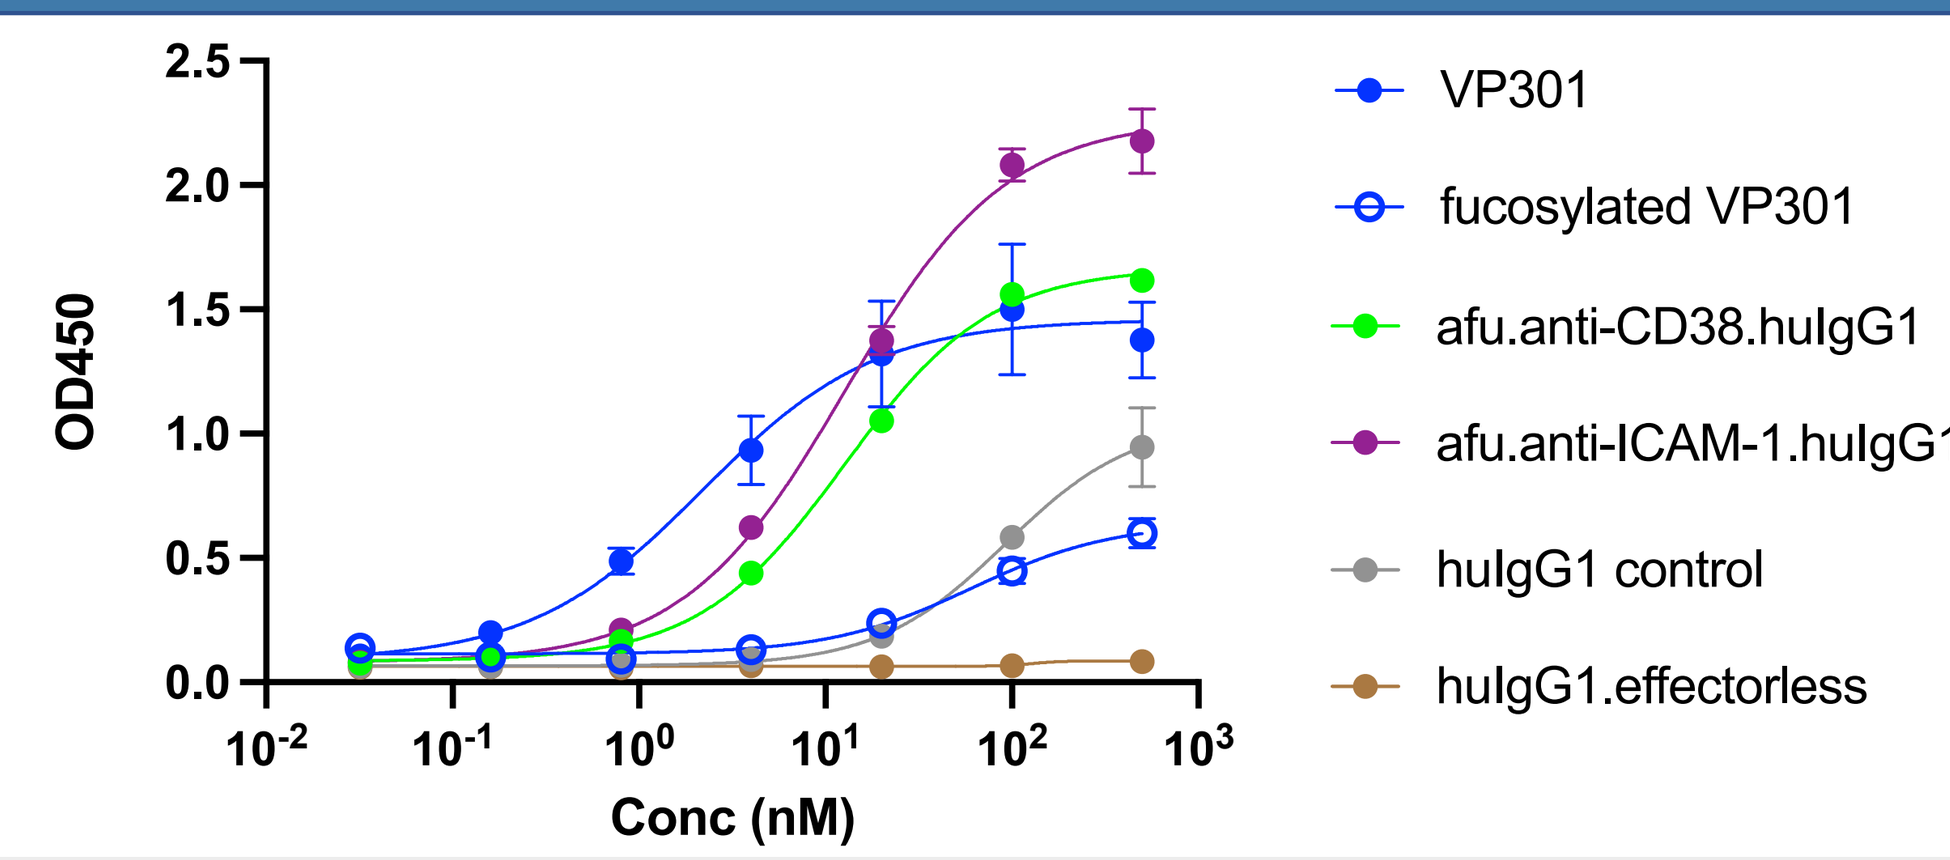
OD450
Conc (nM)
VP301
fucosylated VP301
afu.anti-CD38.huIgG1
afu.anti-ICAM-1.huIgG1
huIgG1 control
huIgG1.effectorless

Fig 5. ELISA binding of VP301 to FcγRIIIa CD16a (V176). VP301 and afucosylated antibodies showed more than 30-fold potency improvement compared to the fucosylated VP301 variant or a native human IgG1 control. As expected, the human IgG1 with effectorless mutation completely abolished FcγRIIIa binding. The afu.anti-CD38.huIgG1 and afu.anti-ICAM-1.huIgG1 represent the afucosylated anti-CD38 antibody and anti-ICAM-1 antibody, respectively.

VP301 Showed Superior Antibody-Dependent Cellular Cytotoxicity (ADCC) Activities

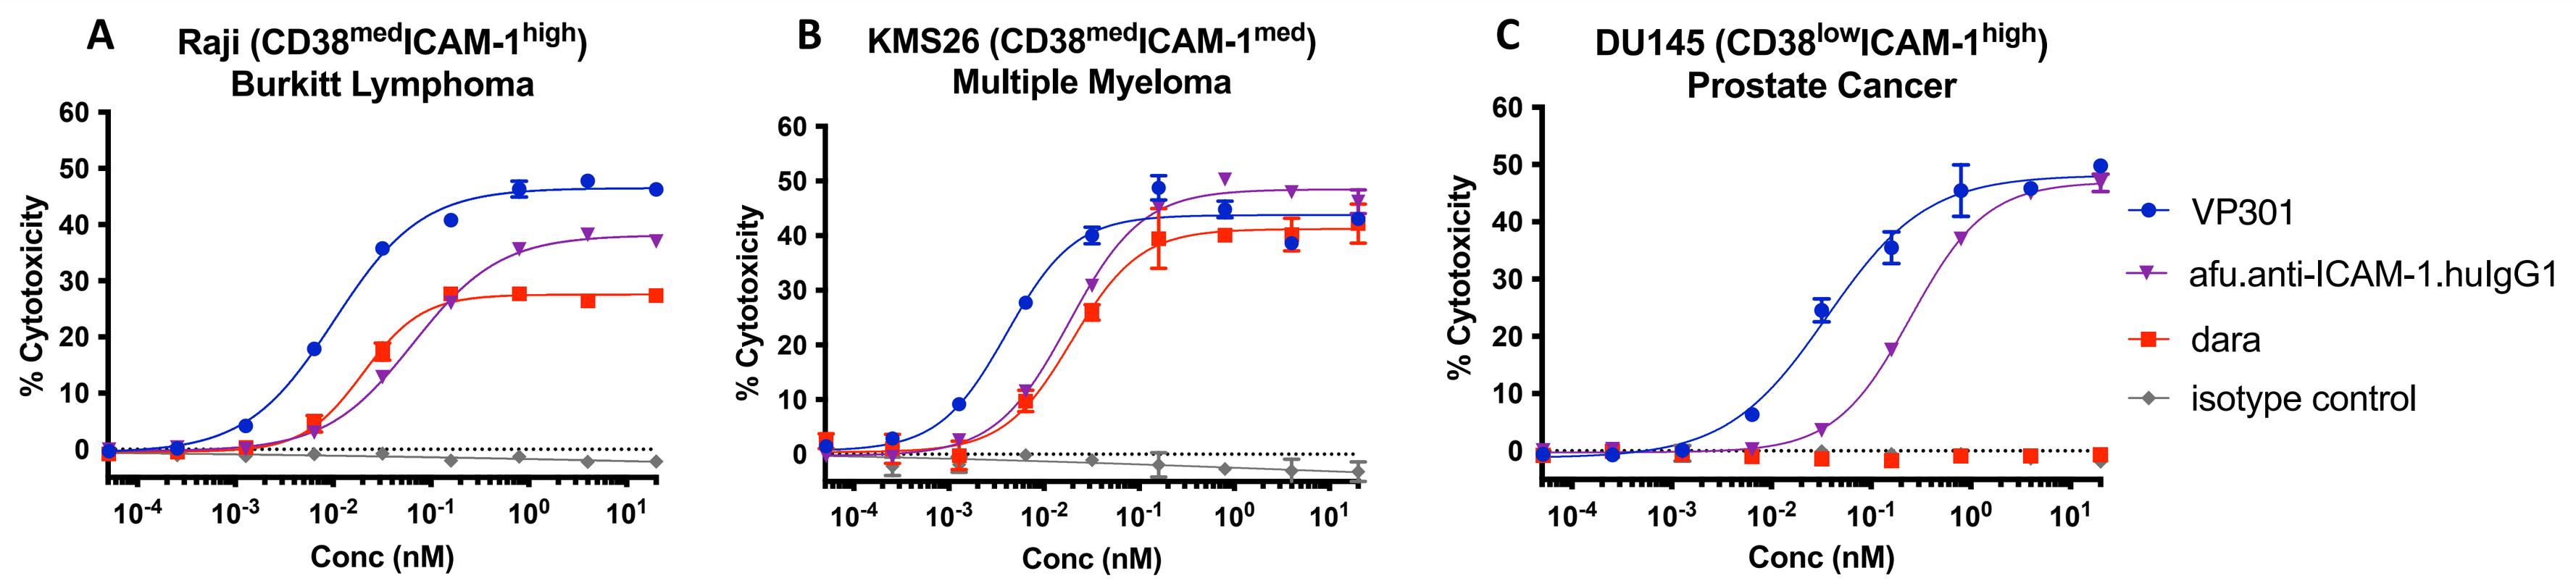
A
Raji (CD38medICAM-1high)
Burkitt Lymphoma
B
KMS26 (CD38medICAM-1med)
Multiple Myeloma
C
DU145 (CD38lowICAM-1high)
Prostate Cancer
% Cytotoxicity
Conc (nM)
VP301
afu.anti-ICAM-1.huIgG1
dara
isotype control

Fig 6. VP301 showed potent *in vitro* ADCC activities on ICAM-1⁺ tumor cells with medium to low CD38 levels, where the benchmark dara has low or minimal effect. ADCC was tested on Raji lymphoma **(A)**, KMS26 MM **(B)** and DU145 prostate **(C)** cancer cells. ADCC activities were measured using the DELFIA time-resolved fluorescence (TRF) cytotoxicity kit (PerkinElmer cat# AD0116) following manufacturer's instruction. Tumor cells were labelled with BATDA. Serially-diluted antibodies were added to assay plate containing human PBMCs and labelled tumor cells at 50:1 effector to target ratio and incubated at 37 °C for 2 hours.

VP301 Showed Reduced *In-Vitro* Natural killer (NK) Cell Fratricide Activity

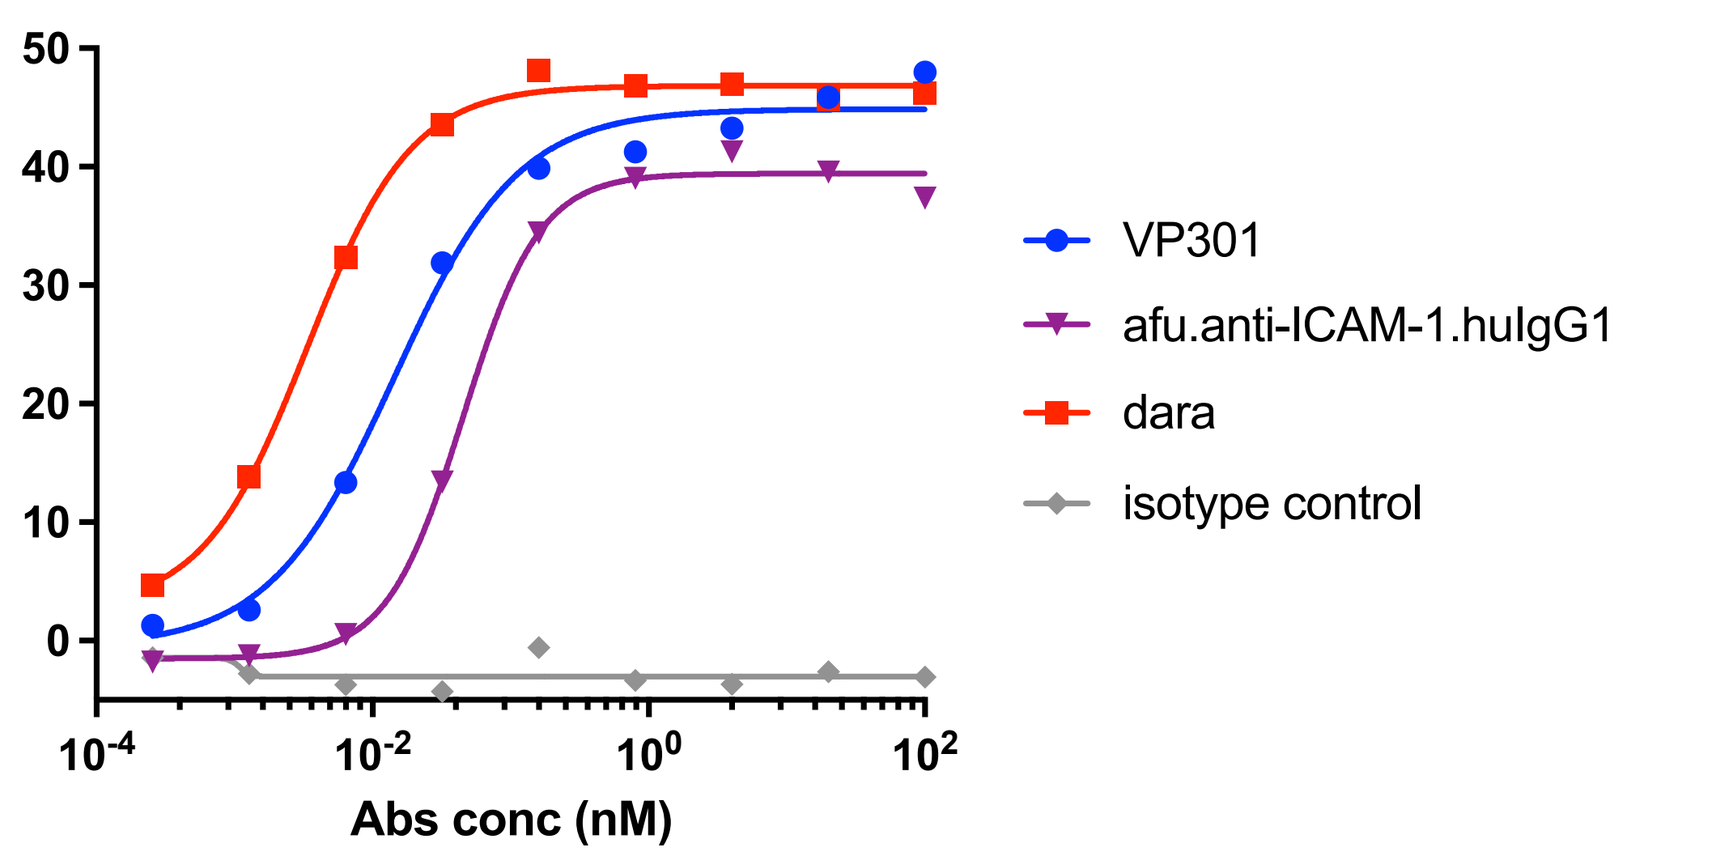
Cytotoxicity%
Abs conc (nM)
VP301
afu.anti-ICAM-1.huIgG1
dara
isotype control

Fig 7. VP301 led to reduced NK92 fratricide compared to the benchmark anti-CD38 antibody dara. The NK92 cells were labelled with BATDA, and the killing activities were measured with the same method as described in Fig 6 except that no effector cell was added.

VP301 Showed Potent Anti-Tumor Activities in Myeloma Cell Line Derived Xenograft (CDX) and Patient Derived Xenograft (PDX) Models

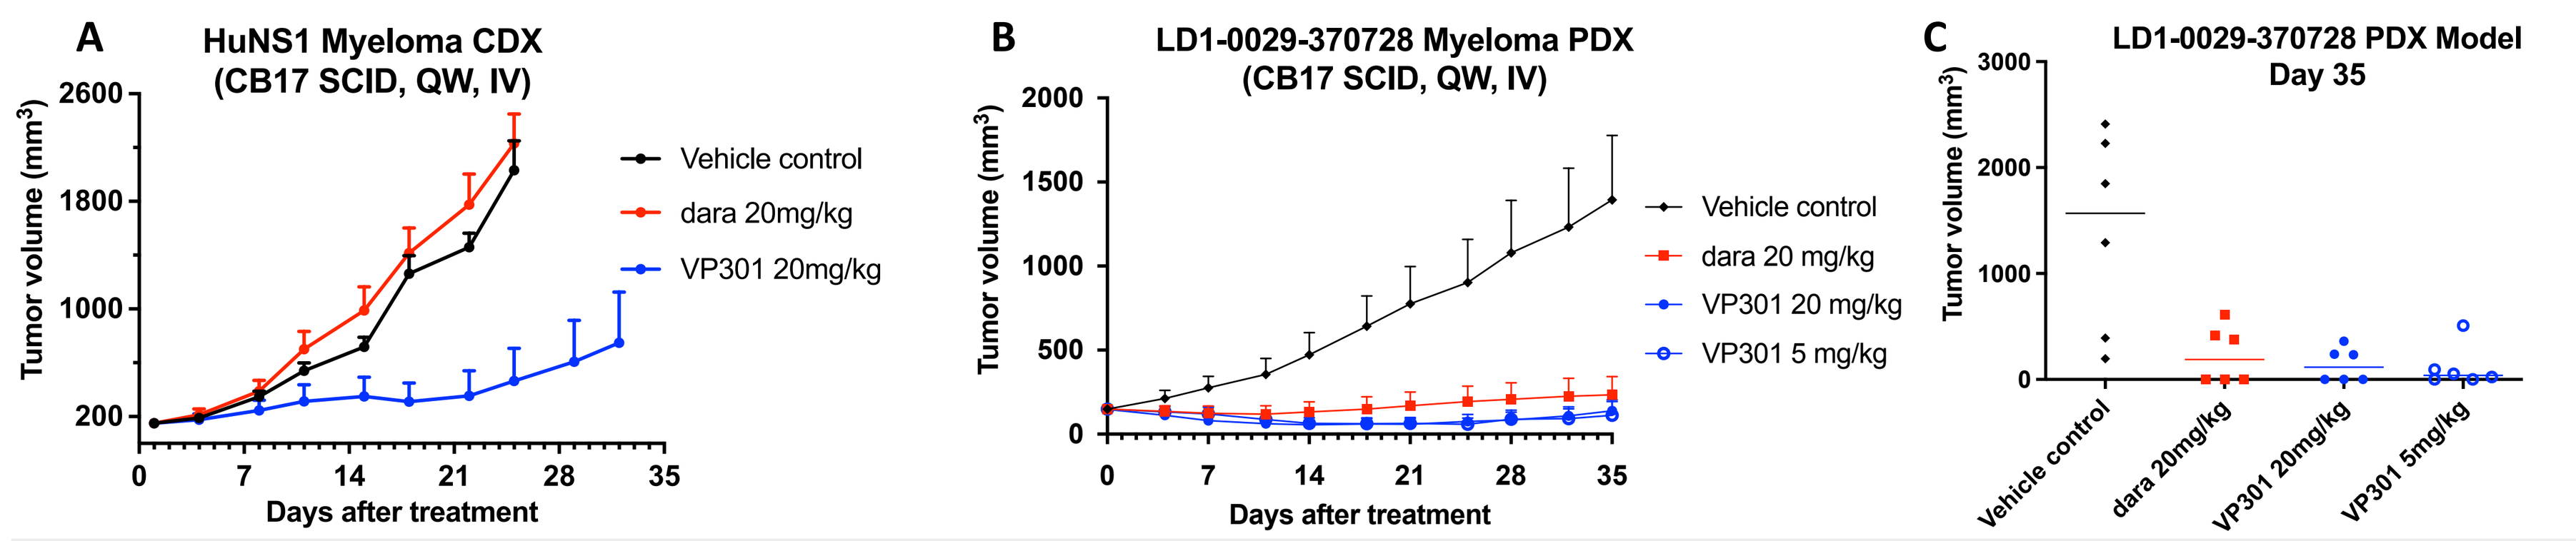
A
HuNS1 Myeloma CDX
(CB17 SCID, QW, IV)
Tumor volume (mm3)
Days after treatment
Vehicle control
dara 20mg/kg
VP301 20mg/kg
B
LD1-0029-370728 Myeloma PDX
(CB17 SCID, QW, IV)
Vehicle control
dara 20 mg/kg
VP301 20 mg/kg
VP301 5 mg/kg
C
LD1-0029-370728 PDX Model
Day 35
Vehicle control
dara 20mg/kg
VP301 20mg/kg
VP301 5mg/kg

Fig 8. The HuNS1 myeloma cells **(A)** were cultured and harvested for tumor inoculation. The PDX model LD1-0029-370728 **(B)** was harvested from tumor bearing mice and cut into small chunks. Female CB17/SCID mice of 6-7 weeks in age were inoculated subcutaneously with myeloma cells or tumor chunks for tumor development. When the mean tumor volume reached approximately 150 mm³, animals were randomized by tumor volumes into 6 animals per group. Test antibodies were administered weekly through bolus tail vein injection at doses mentioned in the graphs. Tumor volume was measured twice weekly. **(C)** Tumor volumes of individual mouse in the LD1-0029-370728 study on day 35 post treatment.

VP301 Showed Potent Anti-Tumor Activities in Both B and T Cell Lymphoma Cell Line Derived Xenograft (CDX) Models

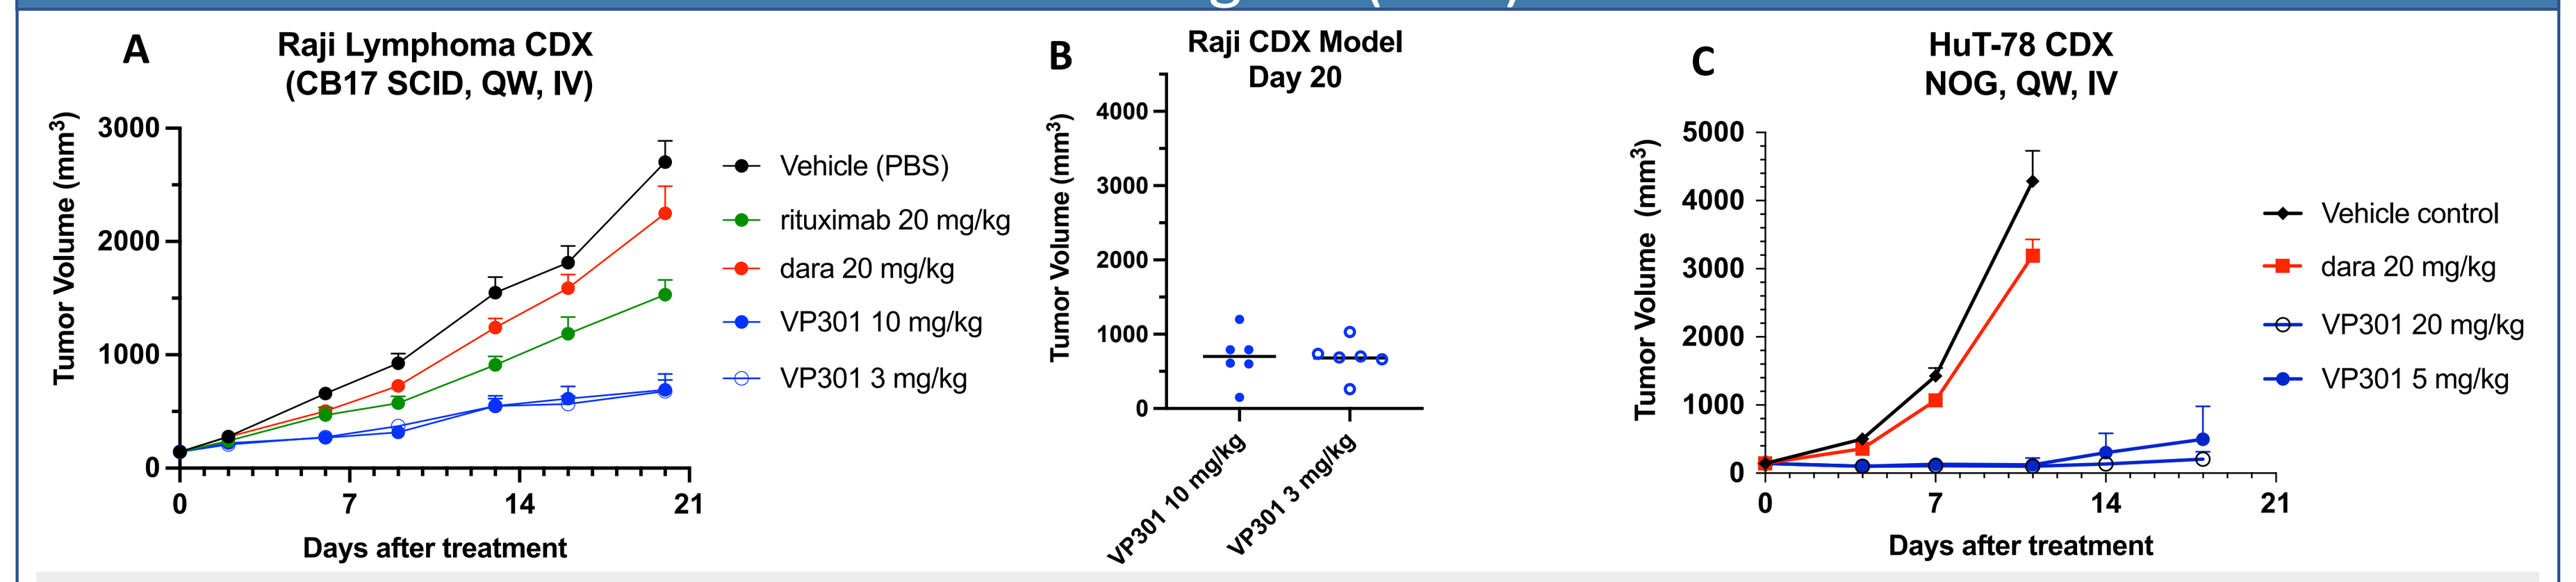
A
Raji Lymphoma CDX
(CB17 SCID, QW, IV)
Tumor Volume (mm3)
Days after treatment
Vehicle (PBS)
rituximab 20 mg/kg
dara 20 mg/kg
VP301 10 mg/kg
VP301 3 mg/kg
B
Raji CDX Model
Day 20
C
HuT-78 CDX
NOG, QW, IV
Vehicle control
dara 20 mg/kg
VP301 20 mg/kg
VP301 5 mg/kg

Fig 9. The B cell lymphoma Raji **(A)** or T cell lymphoma HuT-78 **(C)** cells were cultured and harvested for tumor inoculation. Female CB17/SCID or NOG mice of 6-8 weeks in age were inoculated subcutaneously for tumor development. When the mean tumor volume reached approximately 150 mm³, animals were randomized by tumor volumes into 6 animals per group. Tumor volume was measured twice weekly. **(B)** VP301 3mg/kg and 10mg/kg treatments showed comparable efficacy as shown by tumor volumes of individual mouse in Raji study on day 20 post treatment.

VP301 and Lenalidomide Demonstrated Strong Synergistic Effect in NAMALWA, an Aggressive EBV+ Burkitt Lymphoma

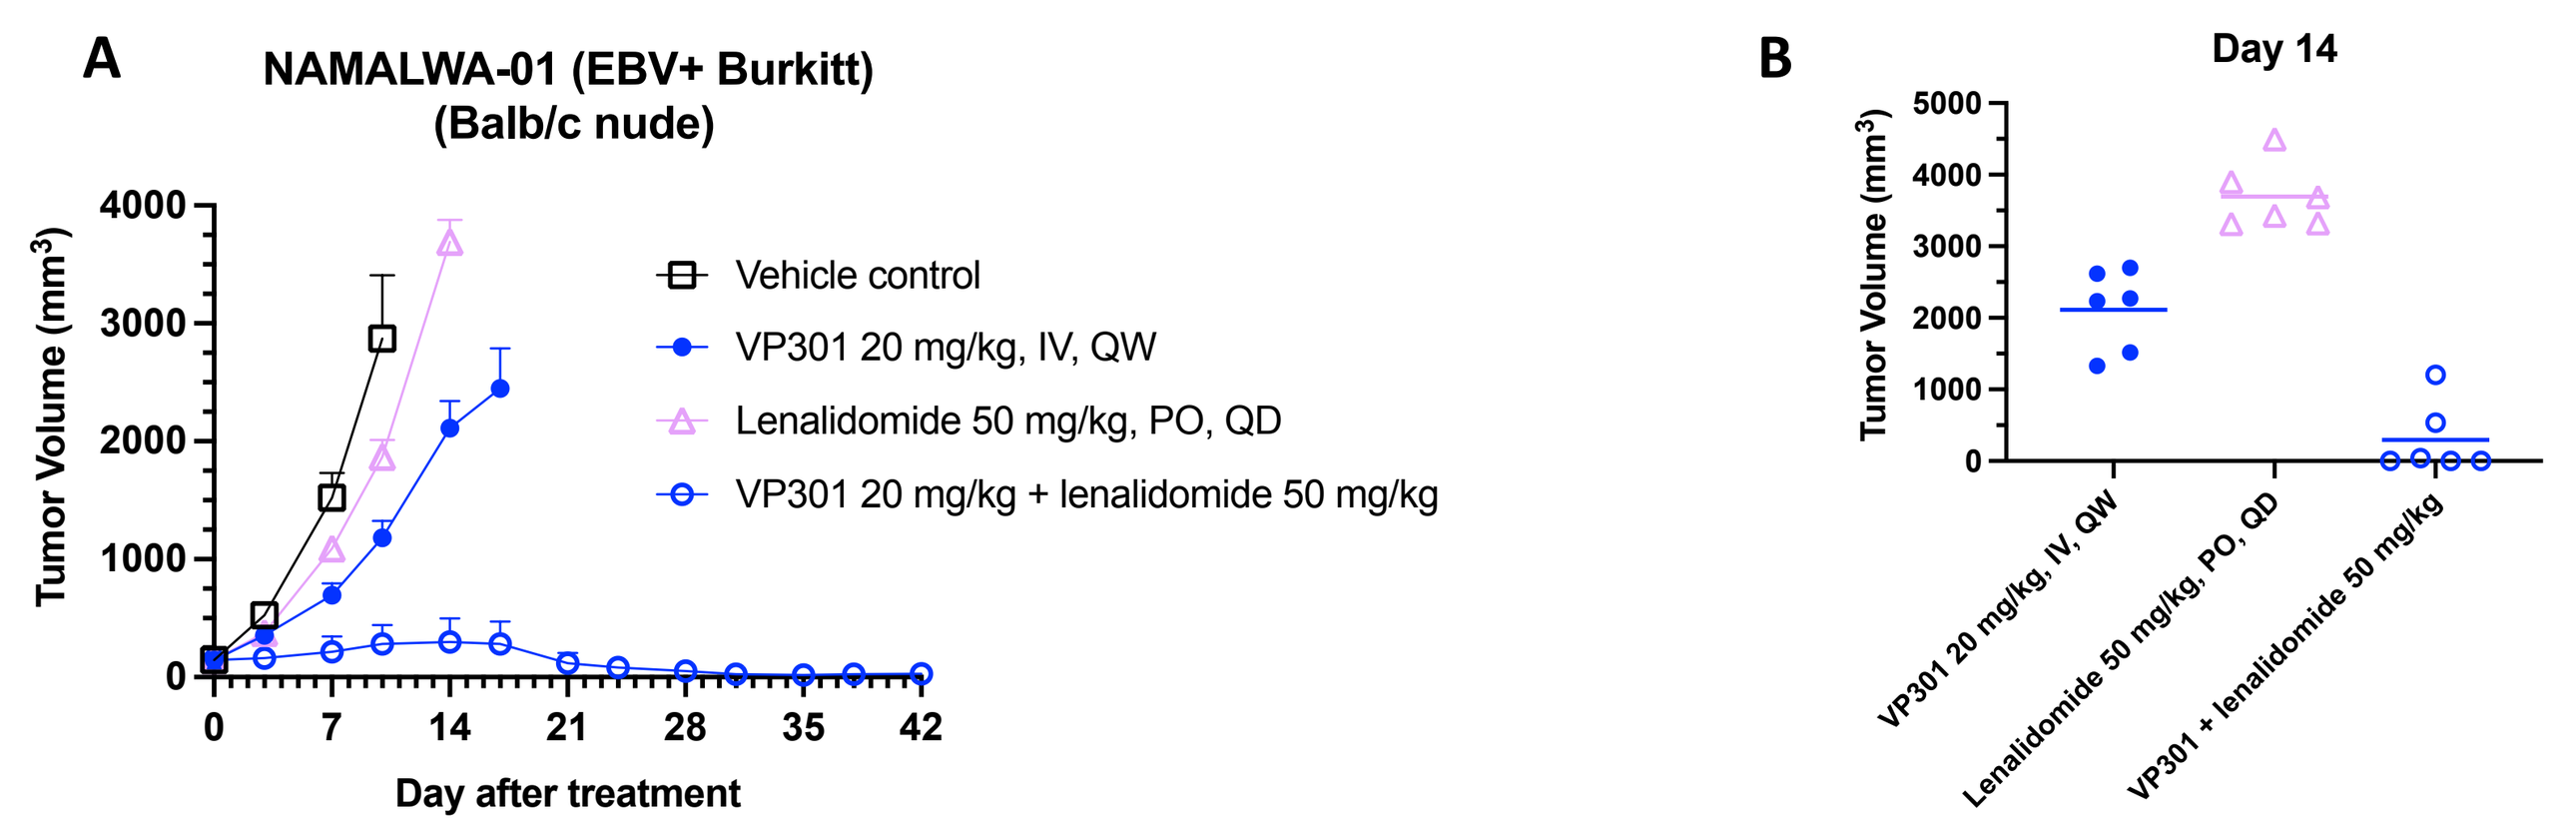
A
NAMALWA-01 (EBV+ Burkitt)
(Balb/c nude)
Tumor Volume (mm3)
Day after treatment
Vehicle control
VP301 20 mg/kg, IV, QW
Lenalidomide 50 mg/kg, PO, QD
VP301 20 mg/kg + lenalidomide 50 mg/kg
B
Day 14
VP301 20 mg/kg, IV, QW
Lenalidomide 50 mg/kg, PO, QD
VP301 + lenalidomide 50 mg/kg

Fig 10. The NAMALWA tumor cells were cultured and harvested for tumor inoculation. Female nude mice of 6-7 weeks in age were inoculated subcutaneously with 10^7 cells in 0.2 mL of PBS supplemented with Matrigel (1:1) for tumor development. When the mean tumor volume reached approximately 150 mm³, animals were randomized by tumor volumes into 6 animals per group. VP301 and lenalidomide were administered at doses mentioned in the graphs. Tumor volume was measured twice weekly. **(A)** Average tumor volumes. **(B)** Tumor volumes of individual mouse on day 14 post treatment.

Conclusions

We developed a novel afucosylated CD38 x ICAM1 bispecific antibody, VP301 that targets unique epitopes on ICAM-1 and CD38. VP301 showed potent *in-vitro* ADCC activities on ICAM1⁺ tumor cells with reduced NK fratricide and RBC binding. The bispecific antibody also showed superior *in-vivo* activities in a variety of multiple myeloma and lymphoma CDX and PDX models. In addition, VP301 demonstrated strong synergistic effect with immunomodulatory drug, lenalidomide. The CD38 x ICAM-1 bispecific antibody VP301 represents a novel approach for treating relapsed/refractory multiple myeloma and lymphoma.

References

1. Lee, N.-K. *et al.* Cell-type specific potent Wnt signaling blockade by bispecific antibody. *Sci. Rep.* **8**, 766 (2018).
2. Park S, Kang S, Chen X, Kim EJ, Kim J, Kim N, et al. Tumor suppression via paclitaxel-loaded drug carriers that target inflammation marker upregulated in tumor vasculature and macrophages. Biomaterials 2013;34:598–605.
3. Sherbenou, D. W. *et al.* Potent Activity of an Anti-ICAM1 Antibody–Drug Conjugate against Multiple Myeloma. *Clin. Cancer Res.* **26**, 6028–6038 (2020).
4. Wichert, S. *et al.* A single-arm, open-label, phase 2 clinical trial evaluating disease response following treatment with BI-505, a human anti-intercellular adhesion molecule-1 monoclonal antibody, in patients with smoldering multiple myeloma. *PLOS ONE* **12**, e0171205 (2017).
5. Yang, Y., Jun, C.-D., Liu, J., Zhang, R., Joachimiak, A., Springer, T.A., and Wang, J. (2004). Structural Basis for Dimerization of ICAM-1 on the Cell Surface. Mol. Cell *14*, 269–276.